Preclinical evaluation of novel immune cell therapies, checkpoint inhibitors, and immune cell engangers in humanized mouse models

A005

Stecklum, Maria[1]; Wulf-Goldenberg, Annika[1]; Brzezicha, Bernadette[1]; Alcaniz, Joshua[1]; Smits, Glenn J.[1]; Walther, Wolfgang[1]; Hoffmann, Jens[1]
[1] Experimental Pharmacology & Oncology GmbH, Berlin, Germany

EUROPÄISCHE UNION
Europäischer Fonds für regionale Entwicklung
Investition in Ihre Zukunft

Background and Aim

- preclinical evaluation of novel immune checkpoint modulators require models with functional immune cells
- in previous experiments, we have demonstrated, that we can use hematopetic stem cells (HSC), peripheral blood mononuclear cells (PBMCs) or subtypes of PBMCs like T or NK cells to establish a humanized immune system on highly immunodeficient mice with functional T, B or NK cells
- by co-transplantation of CDX and PDX, we successfully generated a fully human tumor-immune-cell model in mice
- for orthotopic models of lymphoma or leukemia models, we generated luciferase labeled cell lines to use bioluminescence to follow up on tumor growth during the study
- model have been experimentally validated in preclinical studies with checkpoint inhibitors
- humanized models will be continuously improved by using new mouse strains or optimized cell numbers

Humanized mouse models with immune cells and PDX - schematic overview

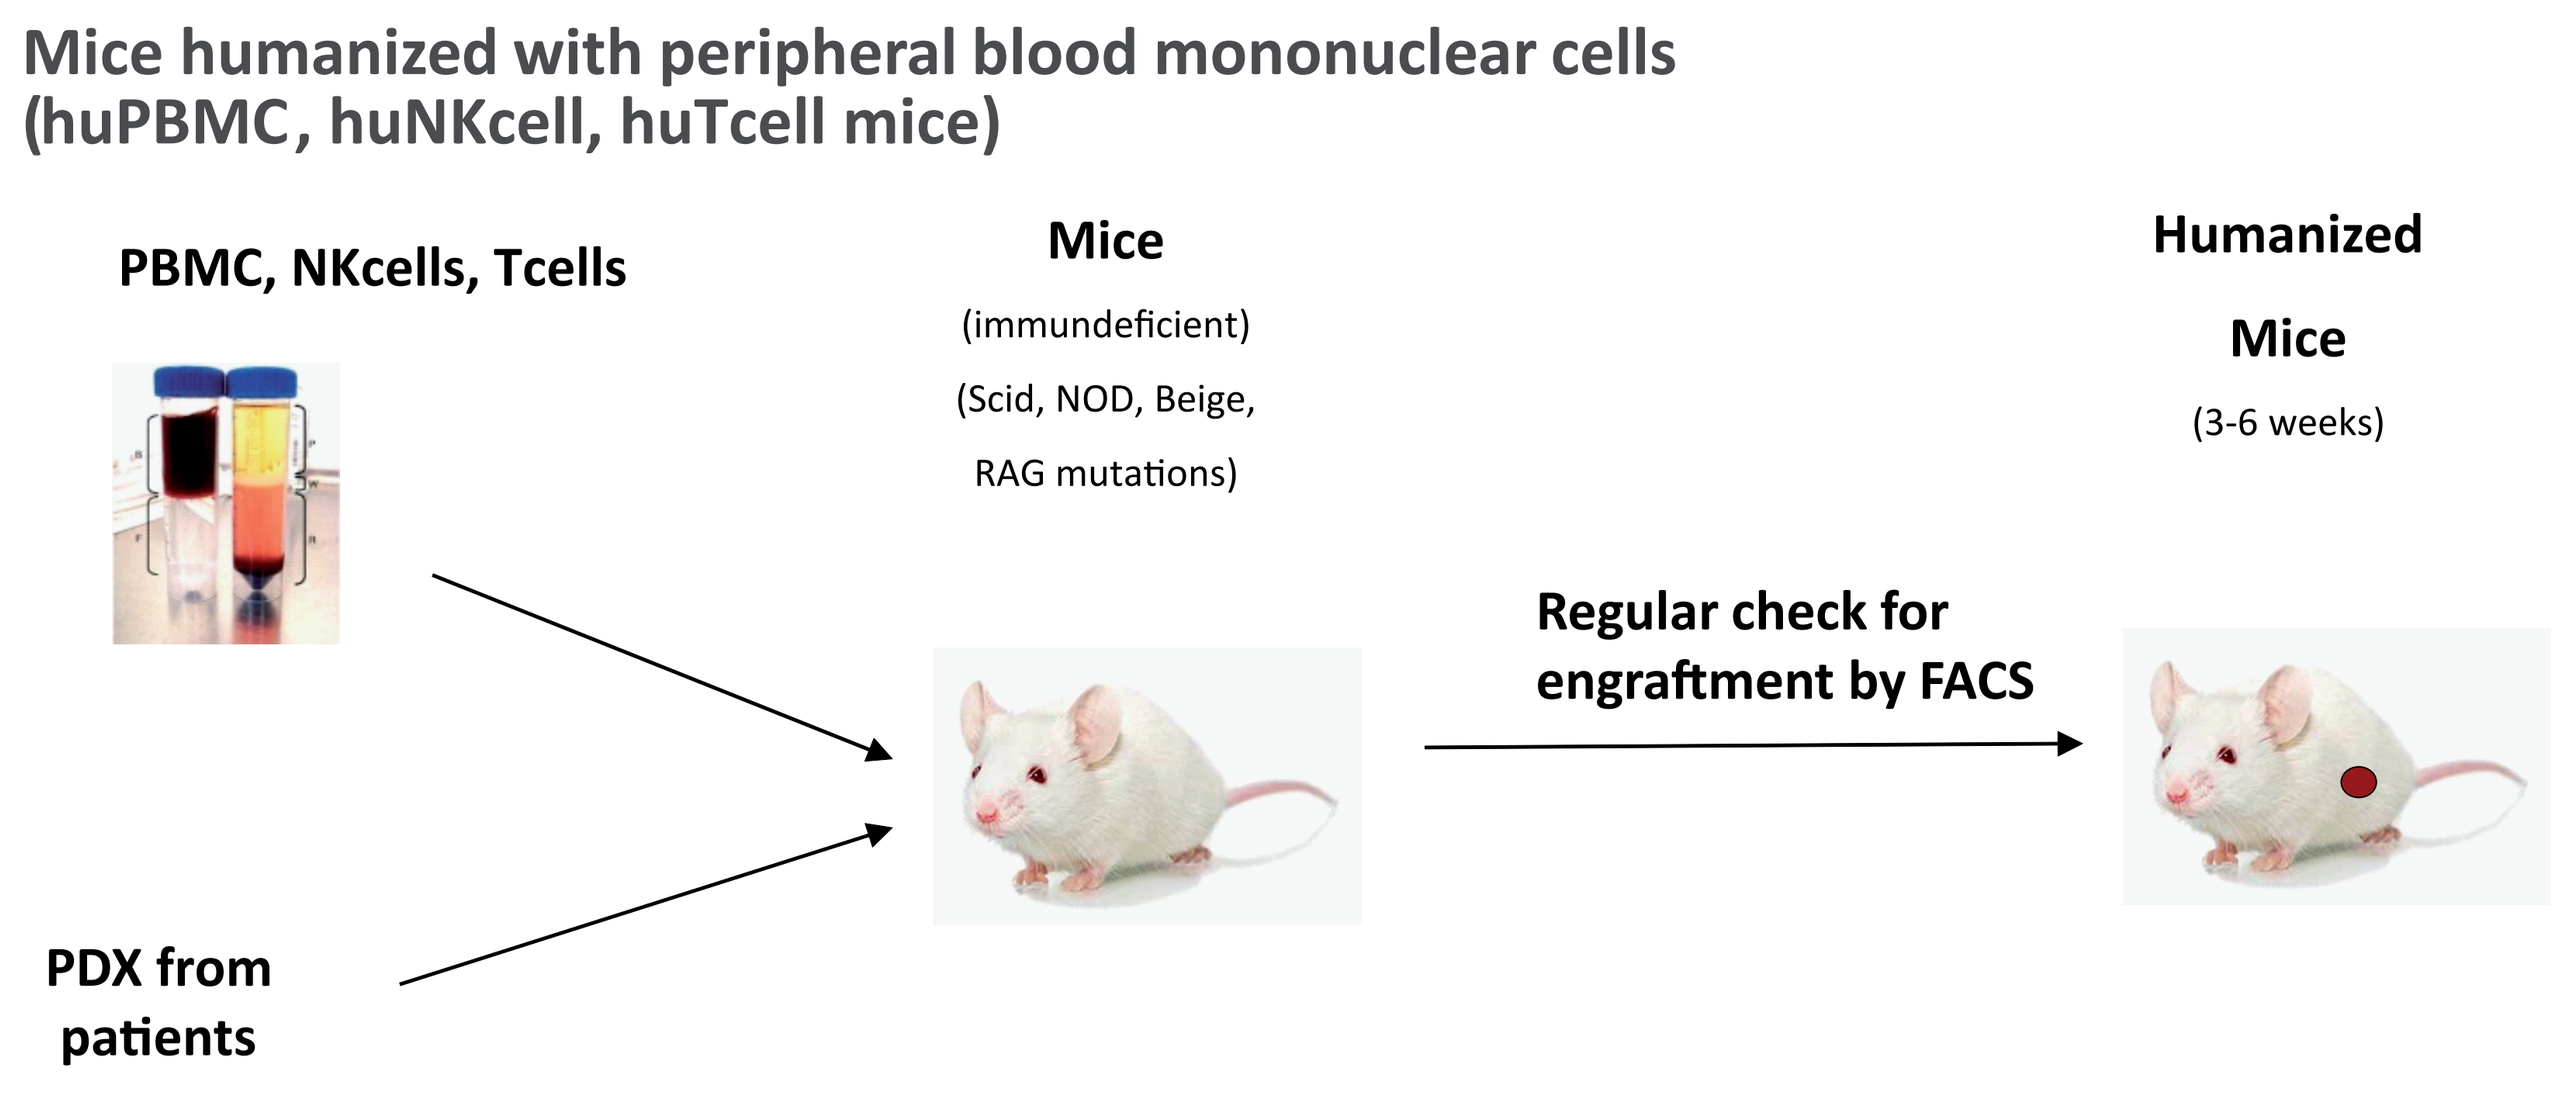
Mice humanized with peripheral blood mononuclear cells (huPBMC, huNKcell, huTcell mice)
PBMC, NKcells, Tcells
Mice
(immundeficient)
(Scid, NOD, Beige, RAG mutations)
PDX from patients
Regular check for engraftment by FACS
Humanized Mice
(3-6 weeks)

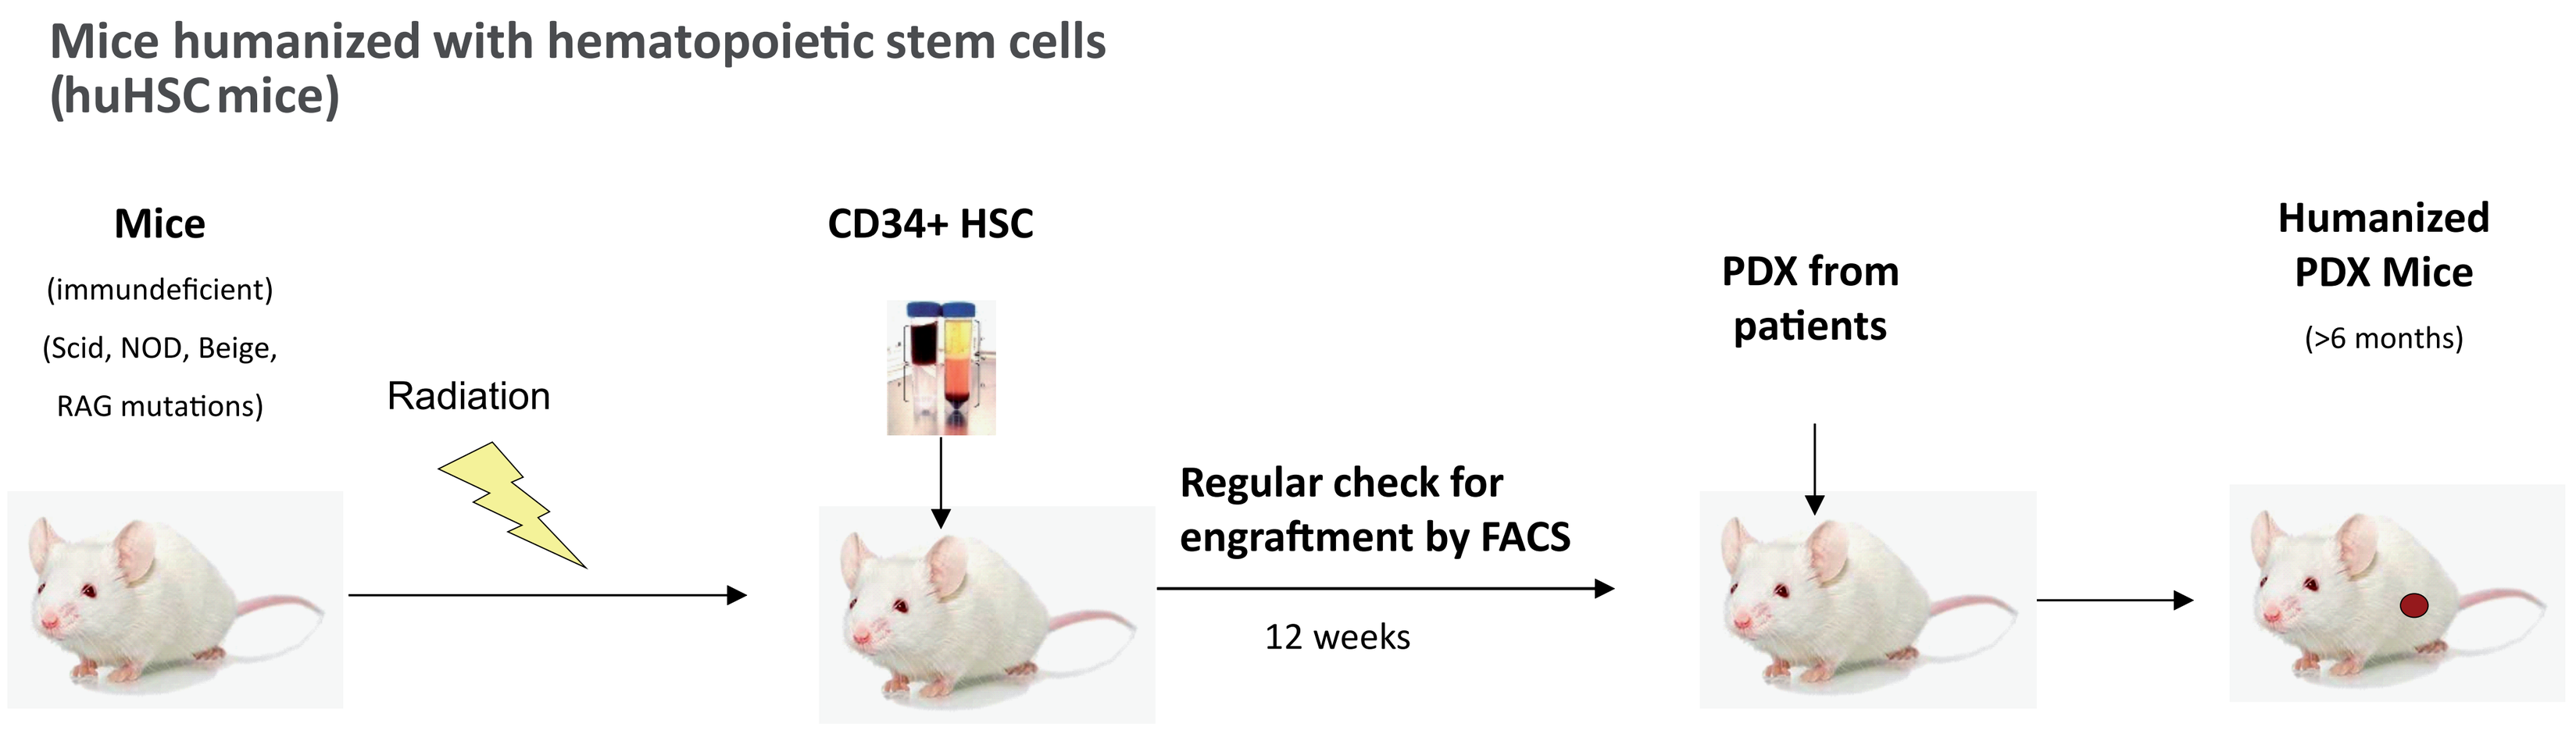
Mice humanized with hematopoietic stem cells (huHSC mice)
Mice
(immundeficient)
(Scid, NOD, Beige, RAG mutations)
Radiation
CD34+ HSC
Regular check for engraftment by FACS
12 weeks
PDX from patients
Humanized PDX Mice
(>6 months)

huPBMC, huNKcell and huTcell mice

- PBMCs, NK or Tcells can be isolated from buffy coats and whole blood samples
- cohort of HLA analyzed donors available from whole blood samples to match with suitable PDX models
- NK and T cells can be expanded in vitro for in vivo studies
- improvement of models with 2nd generation NOG mouse strains (IL15, IL6 and IL2)

huHSC mice

- HSC could reconstitue a humanized immune system in mice
- long-term engraftment could be observed over 14 weeks
- functionality of T cells was determined by inhibition of PD-1 expression through treatment with Nivolumab
- improvement of model with 2nd generation NOG mouse strains (NOG-EXL, NOG-IL6 and supplementation of cytokines

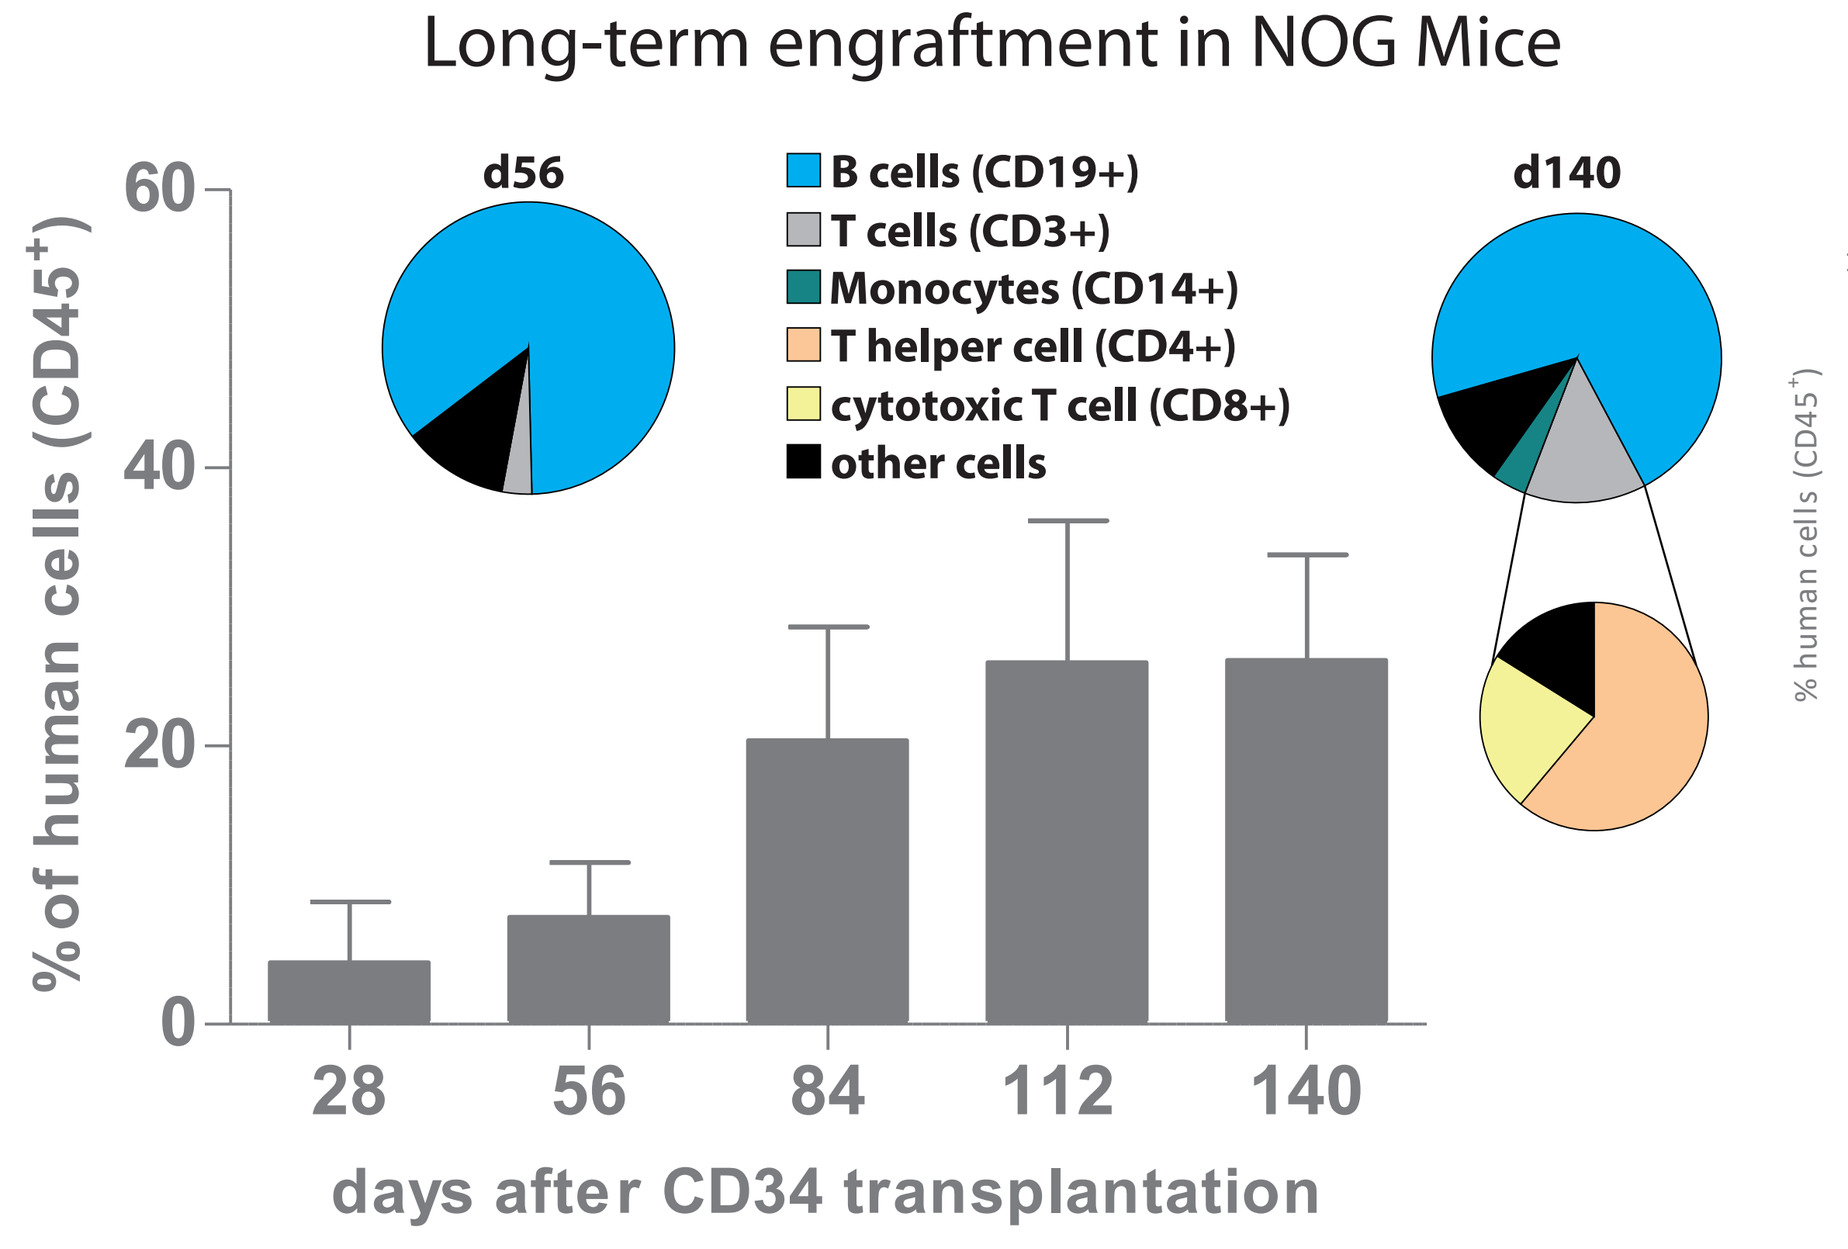
Long-term engraftment in NOG Mice
% of human cells (CD45+)
d56
d140
B cells (CD19+)
T cells (CD3+)
Monocytes (CD14+)
T helper cell (CD4+)
cytotoxic T cell (CD8+)
other cells
28
56
84
112
140
days after CD34 transplantation

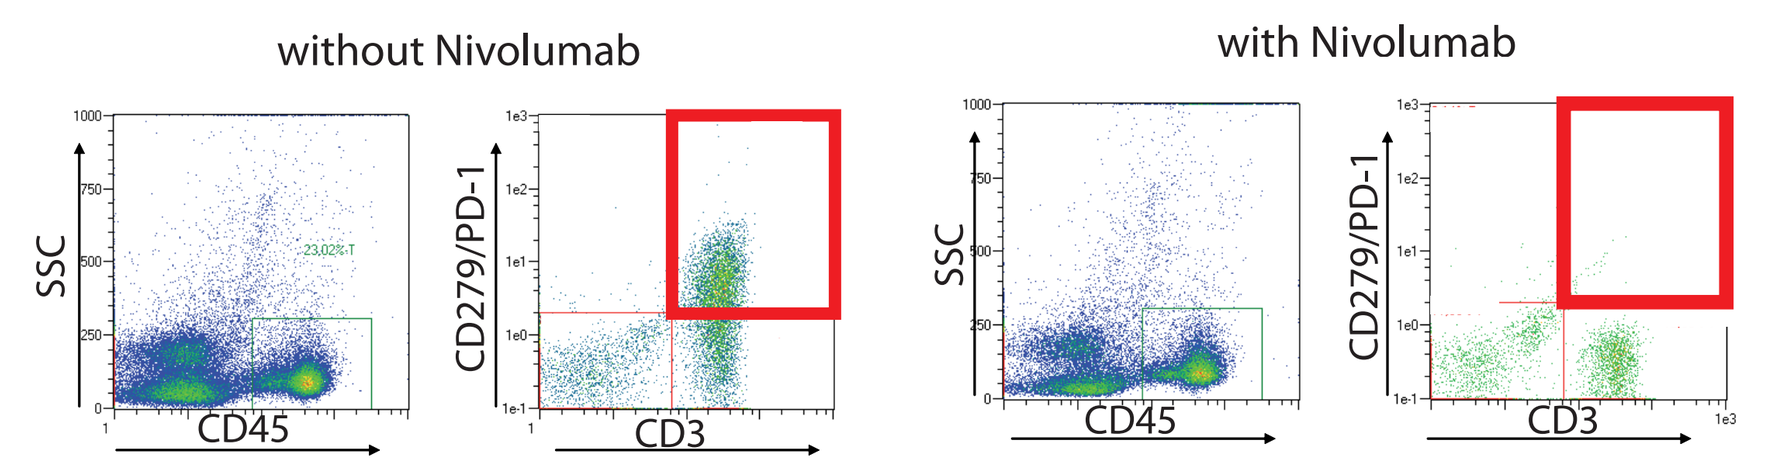
without Nivolumab
with Nivolumab
SSC
CD45
CD279/PD-1
CD3

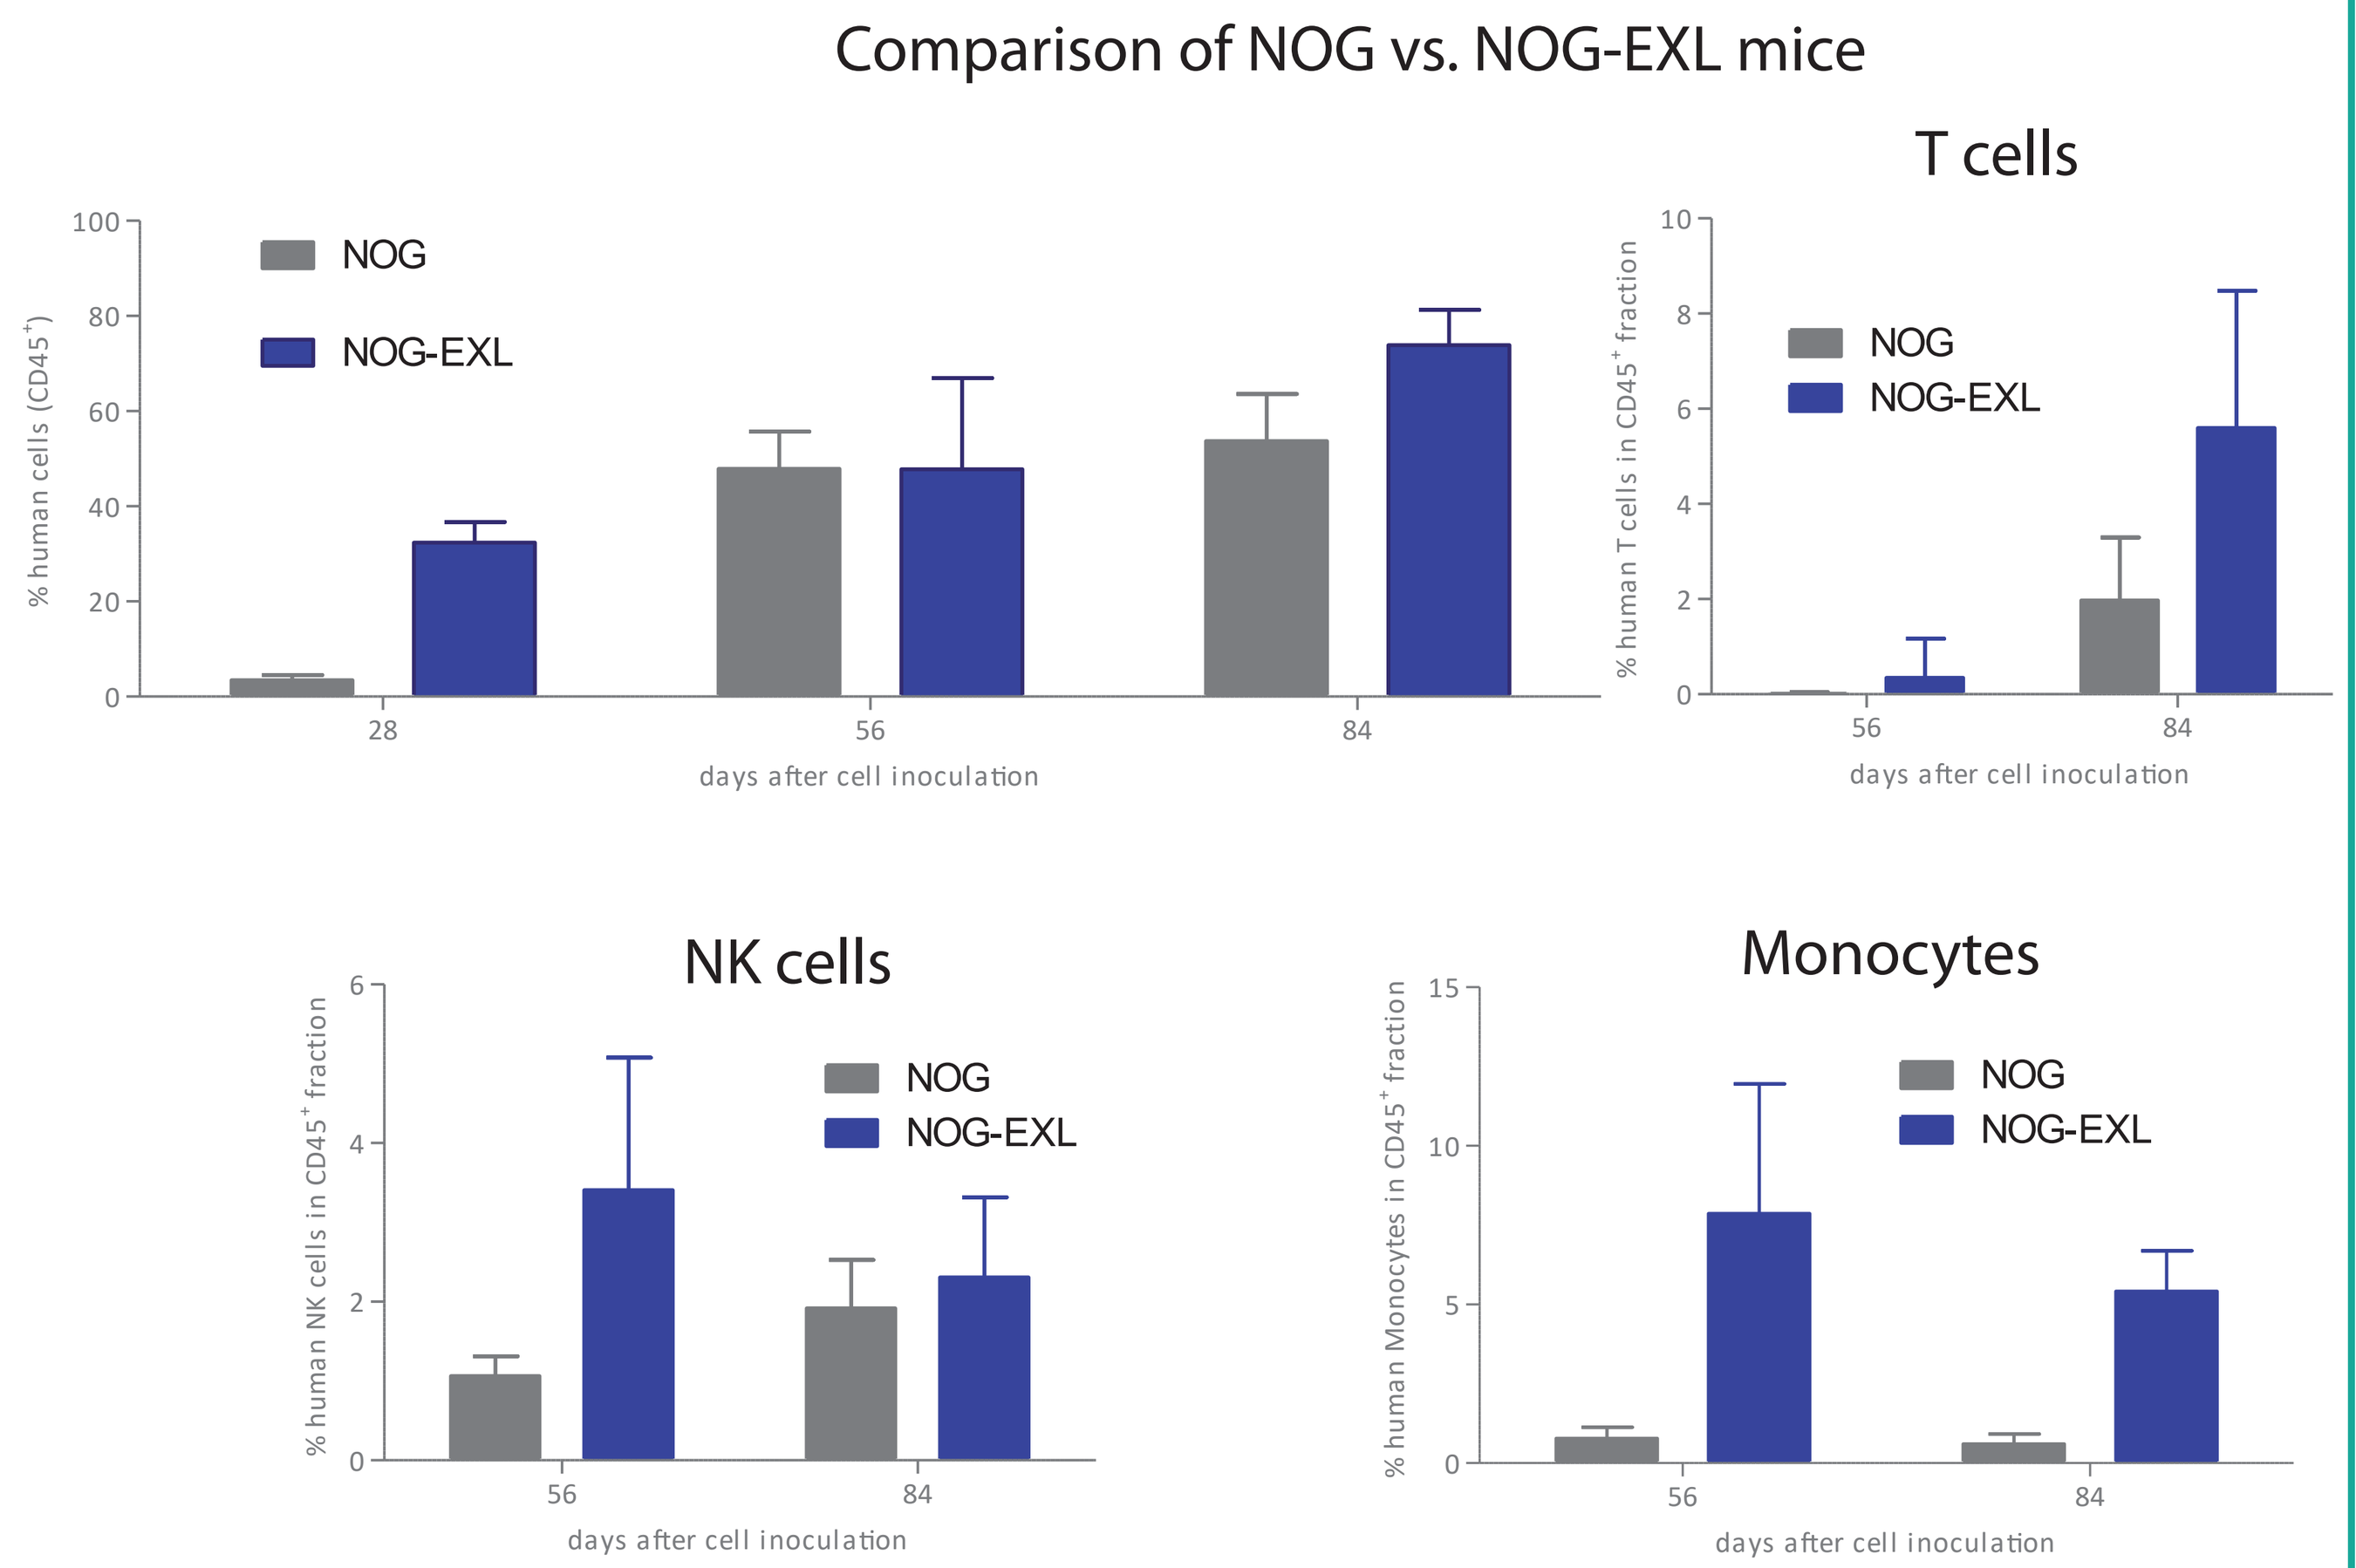
Comparison of NOG vs. NOG-EXL mice
% human cells (CD45+)
NOG
NOG-EXL
days after cell inoculation
T cells
% human T cells in CD45+ fraction
NK cells
% human NK cells in CD45+ fraction
Monocytes
% human Monocytes in CD45+ fraction

Summary and Outlook

- we successfully established a fully humanized mouse models for immuno-oncology studies by co-transplantation of CDX or PDX and human HSC or immune cells from whole blood (PBMCs, T, or NK cells)
- we observed engraftment of CDX and PDX on most humanized mice, however in some cases it was delayed and seems to be dependent on HLA matching and PD-L1 expression
- we see different therapeutic effects of checkpoint inhibitors like Nivolumab, Pembrolizumabm or Ipilimumab with strong, to minor responders, or non responders
- several CDX and PDX have been investigated in humanized HSC mice, huPBMC, huNK cells and huT cell mice
- comparing tumor growth and checkpoint inhibitor activity in the pancreatic cancer PDX Panc12975 on four different humanized mouse models, humanization with HSC provided best results in comparison to single immune cells
- we successfully established an orthotopic model of lymphoma model Karpas299 to be used for immuno-oncology research, tumor growth can be measured with bioluminescene during the experiment
- we demonstrated in our preclinical studies eligibility of the humanized models for peclinical research in tumor immunology, evaluation of new therapies and combinations, as well as the identification and validation of biomarkers for immune therapy
- combination therapies with radiation and using mouse strains improving engraftment of HSC (NOG-EXL mice) and immune cells (NOG-IL-15 mice) are under investigation
- furthermore, these novel models have been successful used for the preclinical evaluation of new bispecific immune cell engagers (BITE) and cell therapies (CAR T cells)

Evaluation of immun therapies with CDX models on humanized mouse models

human HSC mice

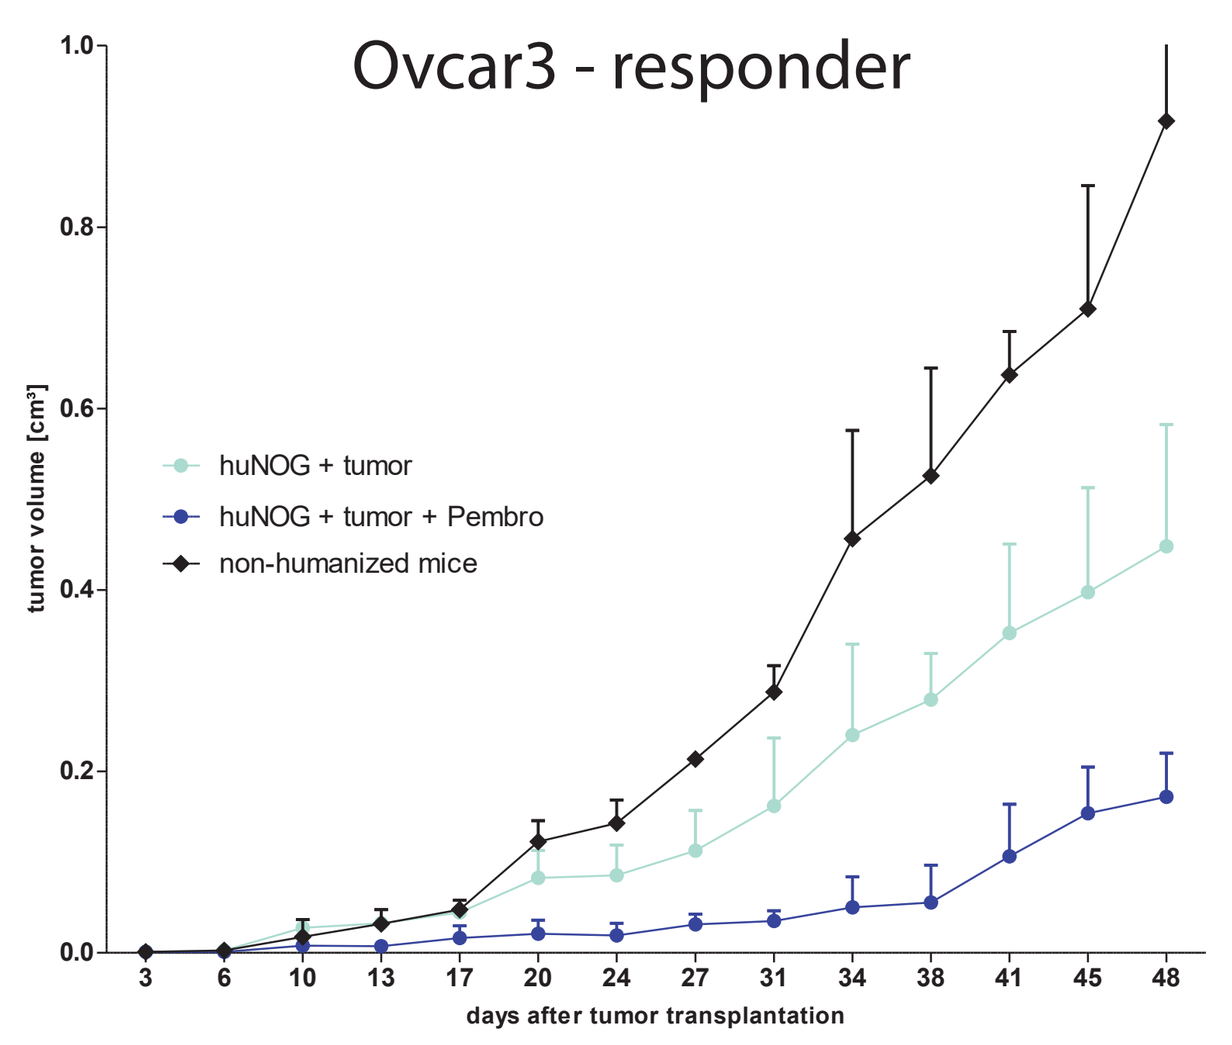
Ovcar3 - responder
tumor volume [cm³]
huNOG + tumor
huNOG + tumor + Pembro
non-humanized mice
days after tumor transplantation

- delayed growth of Ovcar3 CDX on humanized HSC mice, distinct efficacy of checkpoint inhibitor Nivolumab could be shown

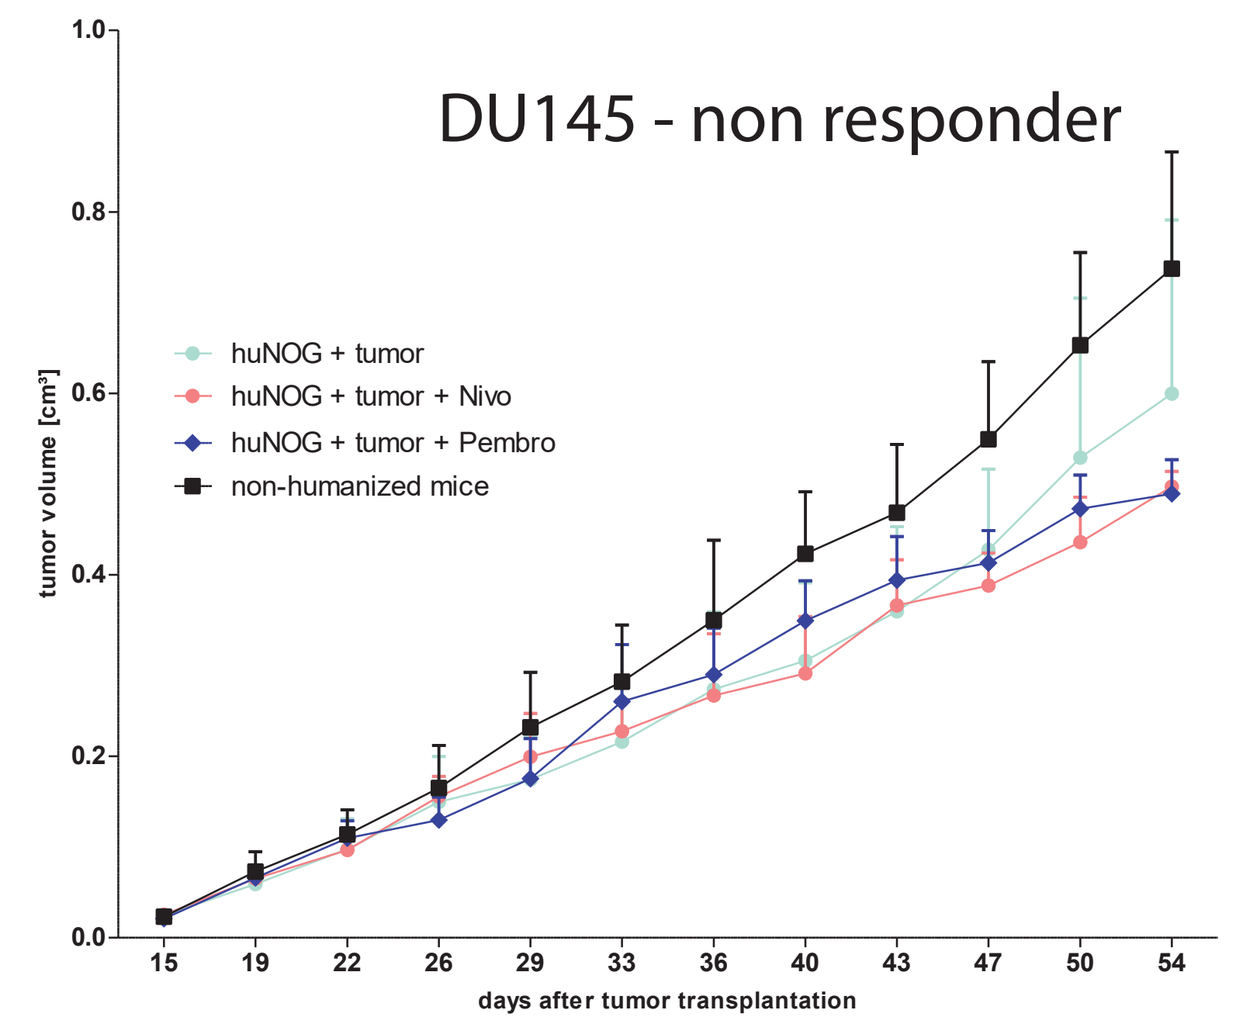
DU145 - non responder
tumor volume [cm³]
huNOG + tumor
huNOG + tumor + Nivo
huNOG + tumor + Pembro
non-humanized mice
days after tumor transplantation

- comparable growth of DU145 CDX on humanized HSC mice, no effects of checkpoint inhibitor Nivolumab and Pembrolizumab could be shown

human PBMC mice

Karpas299/Luc - responder

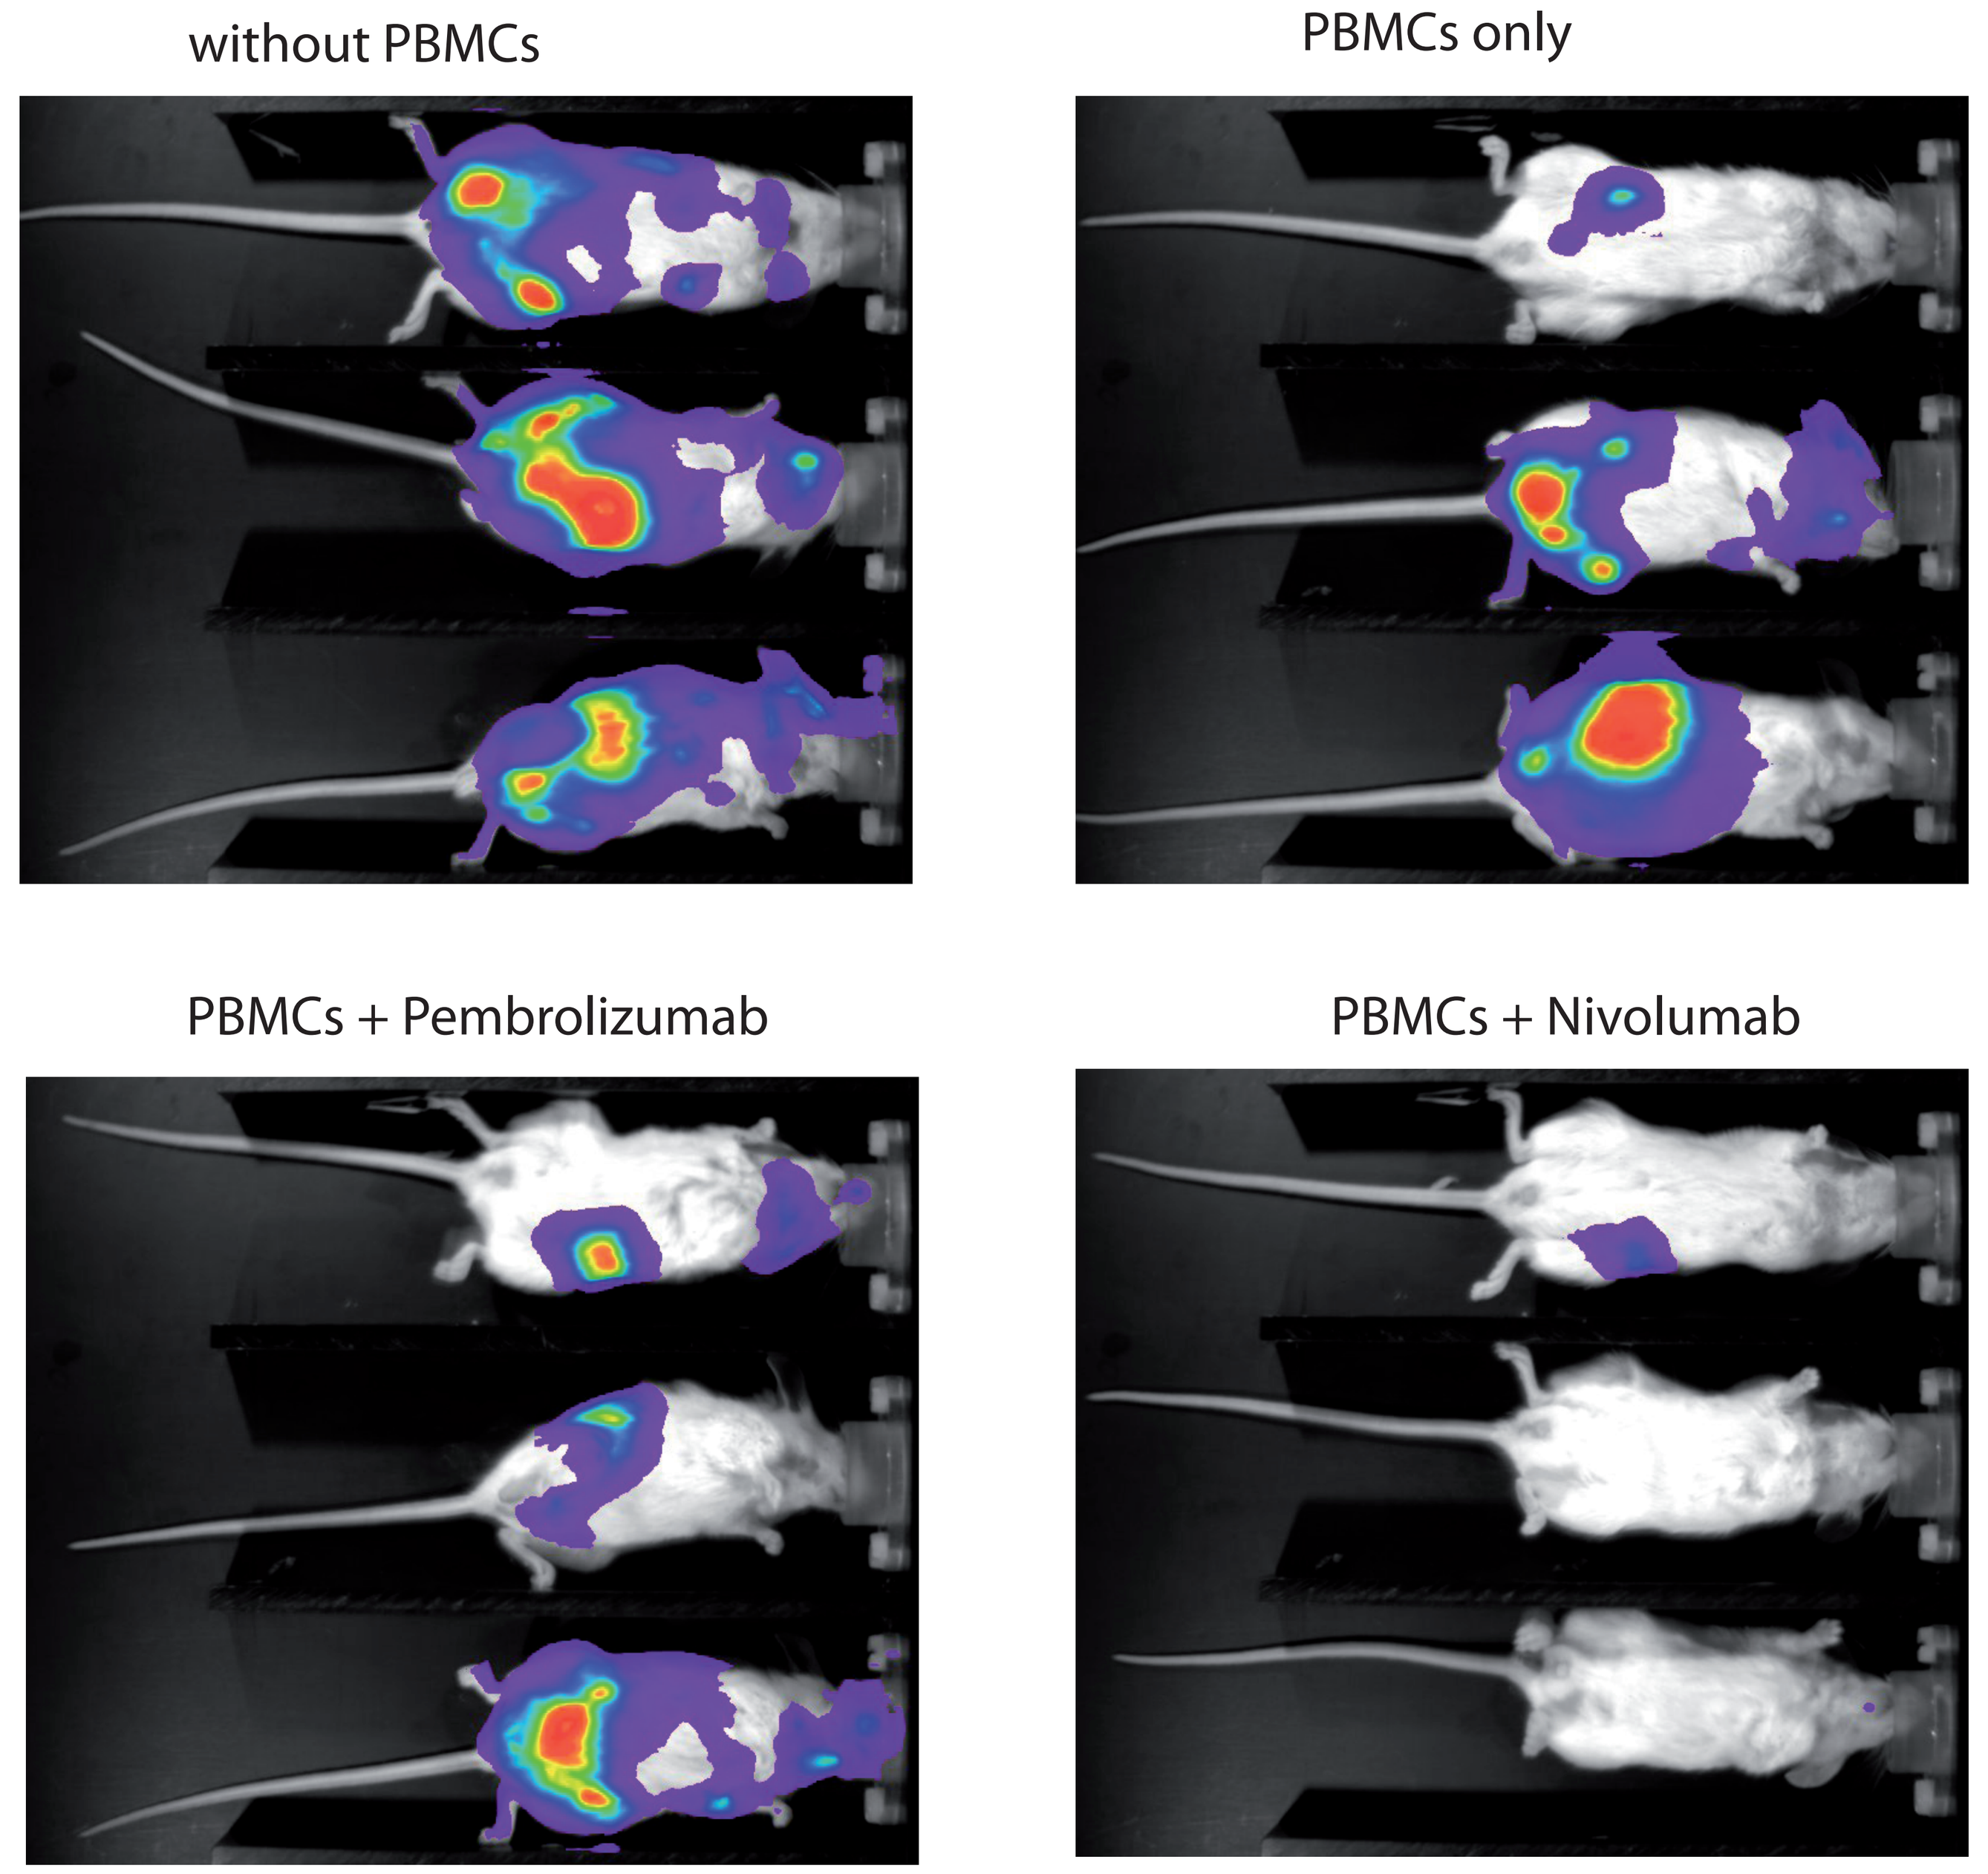
without PBMCs
PBMCs only
PBMCs + Pembrolizumab
PBMCs + Nivolumab

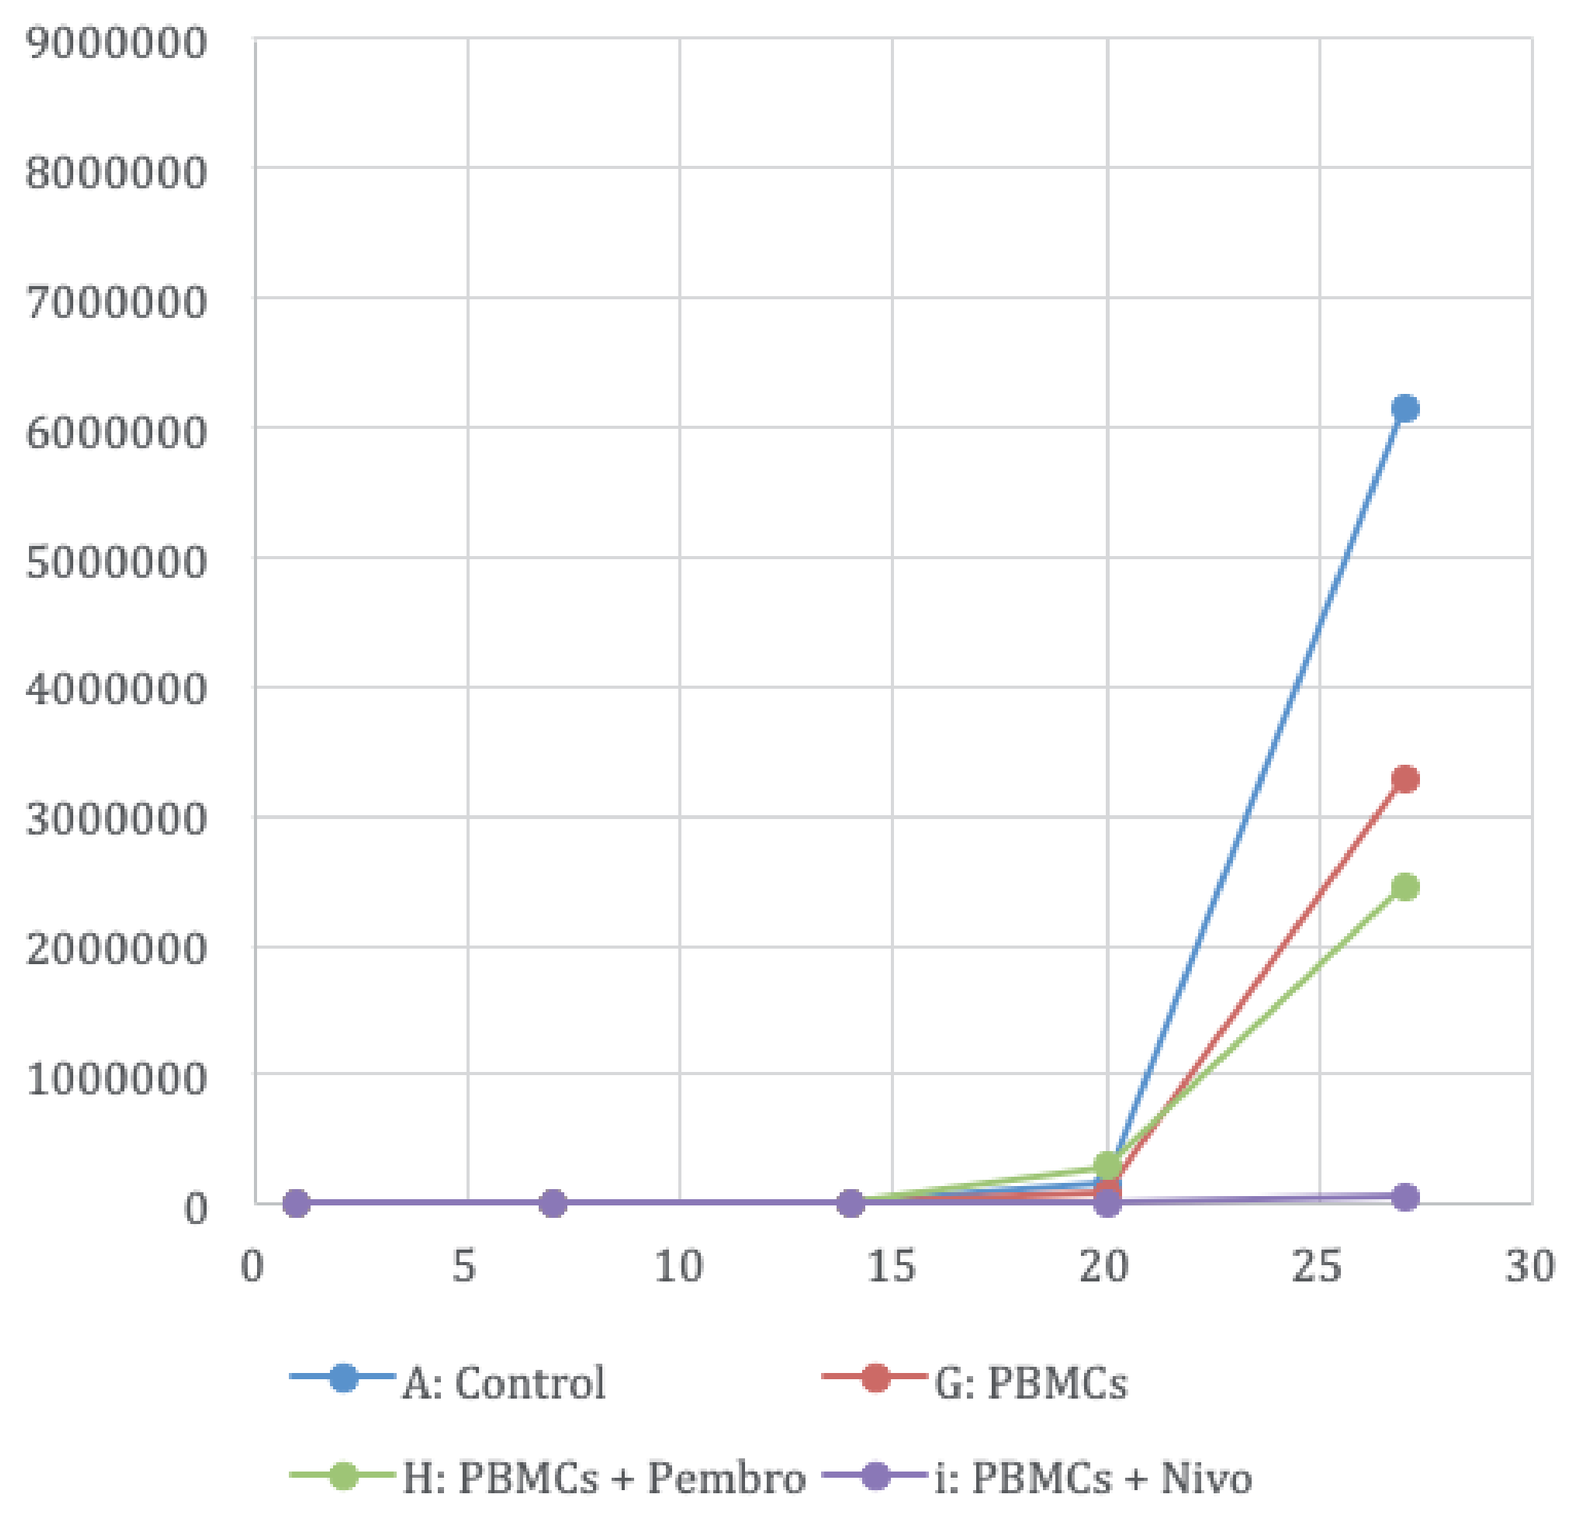
A: Control
G: PBMCs
H: PBMCs + Pembro
i: PBMCs + Nivo

- comparable tumor growth of with and without PBMCs
- tumor growth delay can be induced by treatment with checkpoint inhibitors
- Nivolumab induced significant tumor growth reduction in comparison to Pembrolizumab

Evaluation of immuno therapies with PDX model Panc12975 on four different humanized mouse models

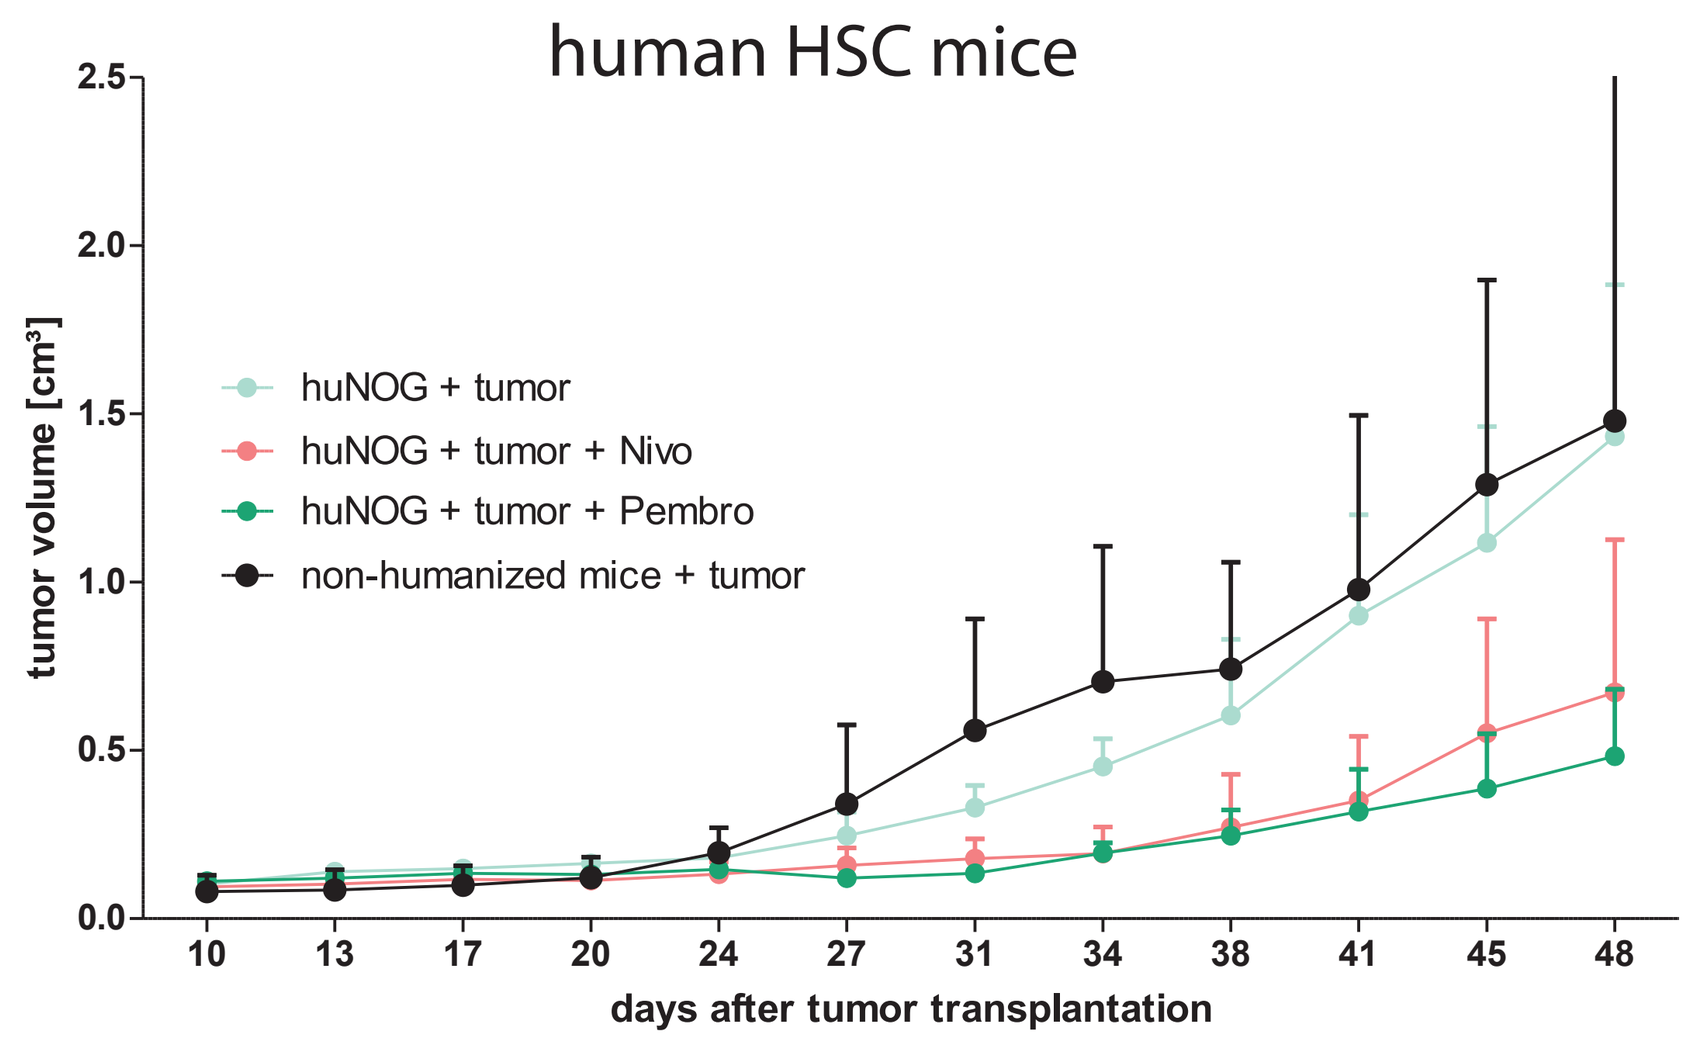
human HSC mice
tumor volume [cm³]
huNOG + tumor
huNOG + tumor + Nivo
huNOG + tumor + Pembro
non-humanized mice + tumor
days after tumor transplantation

- comparable engraftment of Panc12975 on huHSC and non-humanized mice, efficacy of checkpoint inhibitors Nivolumab and Pembrolizumab could be shown in Panc12975 PDX model

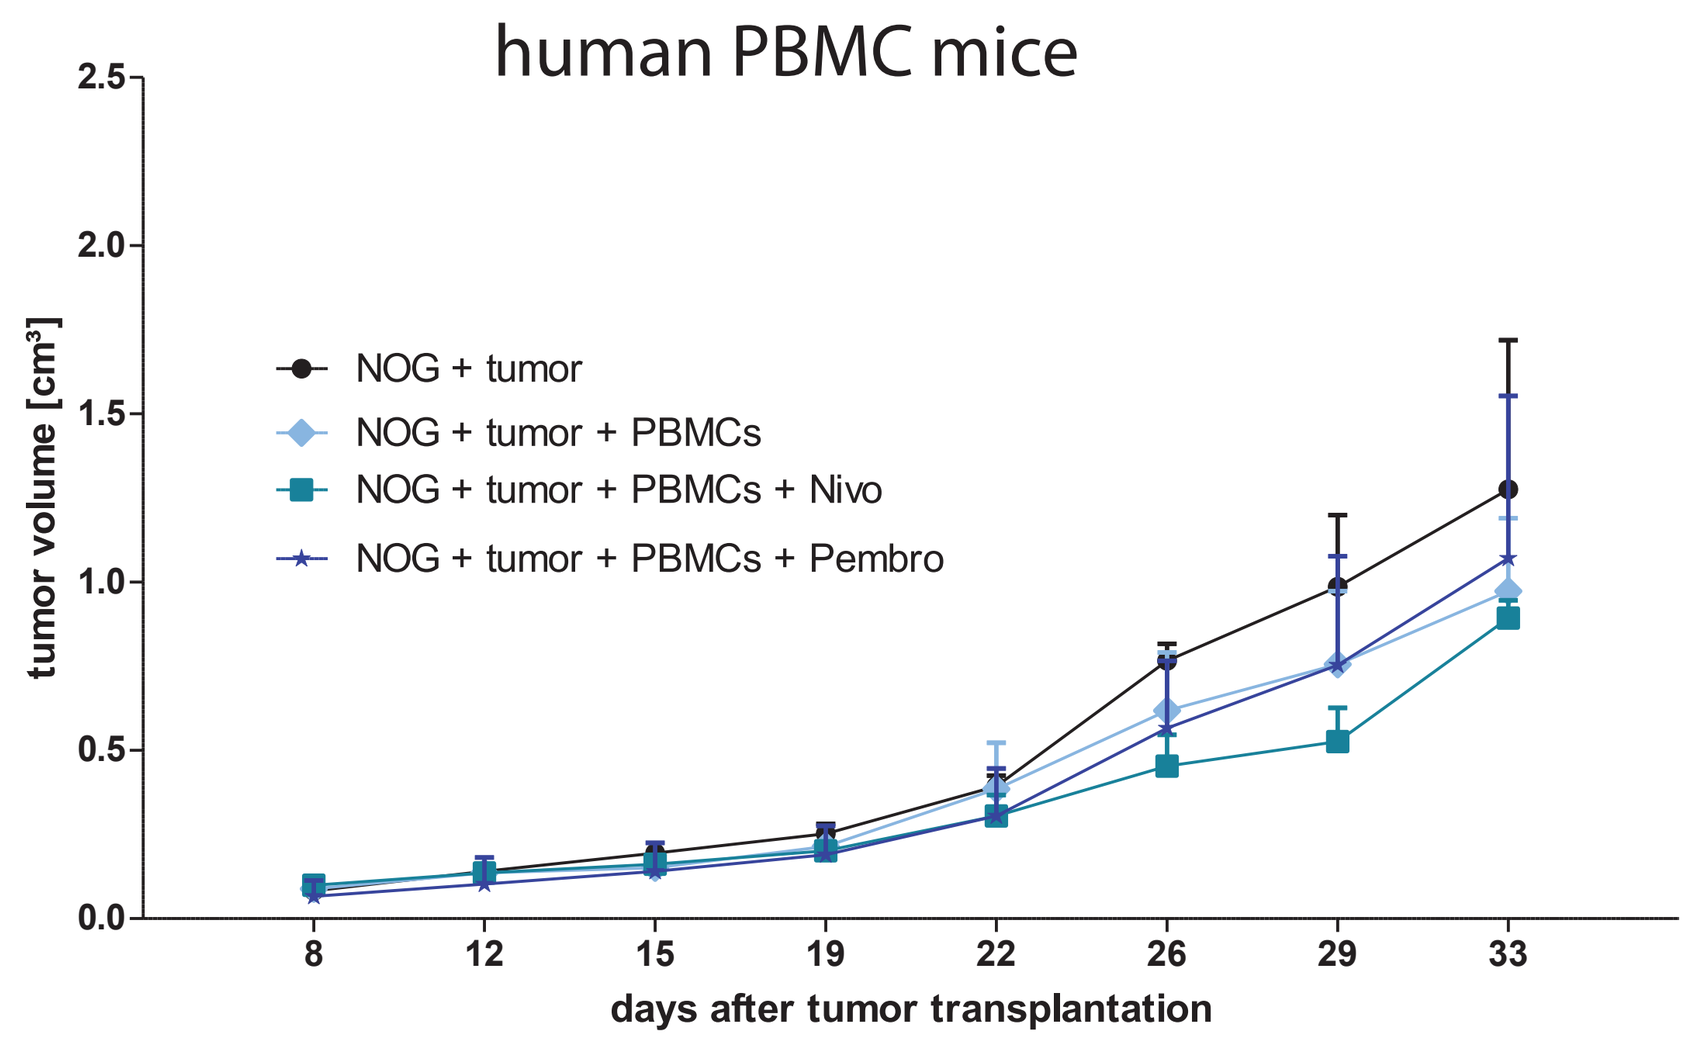
human PBMC mice
tumor volume [cm³]
NOG + tumor
NOG + tumor + PBMCs
NOG + tumor + PBMCs + Nivo
NOG + tumor + PBMCs + Pembro
days after tumor transplantation

- comparable growth of Panc12975 on mice with and without PBMC humanization, no effects of checkpoint inhibitors

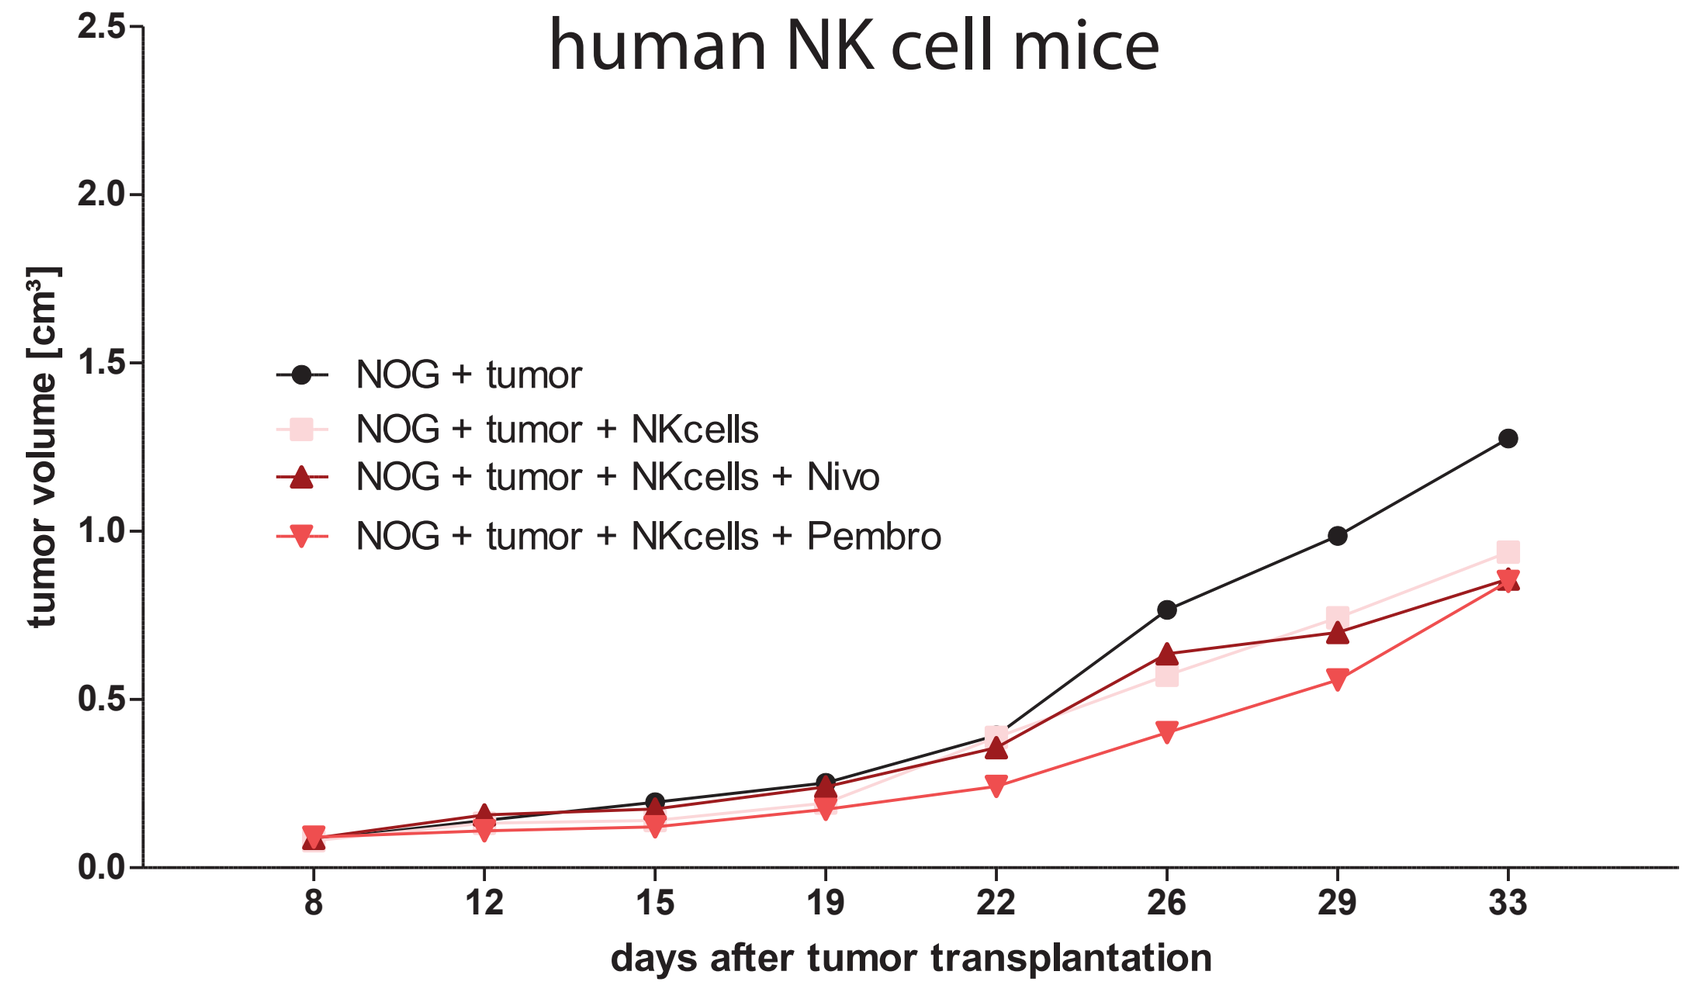
human NK cell mice
tumor volume [cm³]
NOG + tumor
NOG + tumor + NKcells
NOG + tumor + NKcells + Nivo
NOG + tumor + NKcells + Pembro
days after tumor transplantation

- comparable growth of Panc12975 on mice with and without NK cell humanization, no effects of checkpoint inhibitors

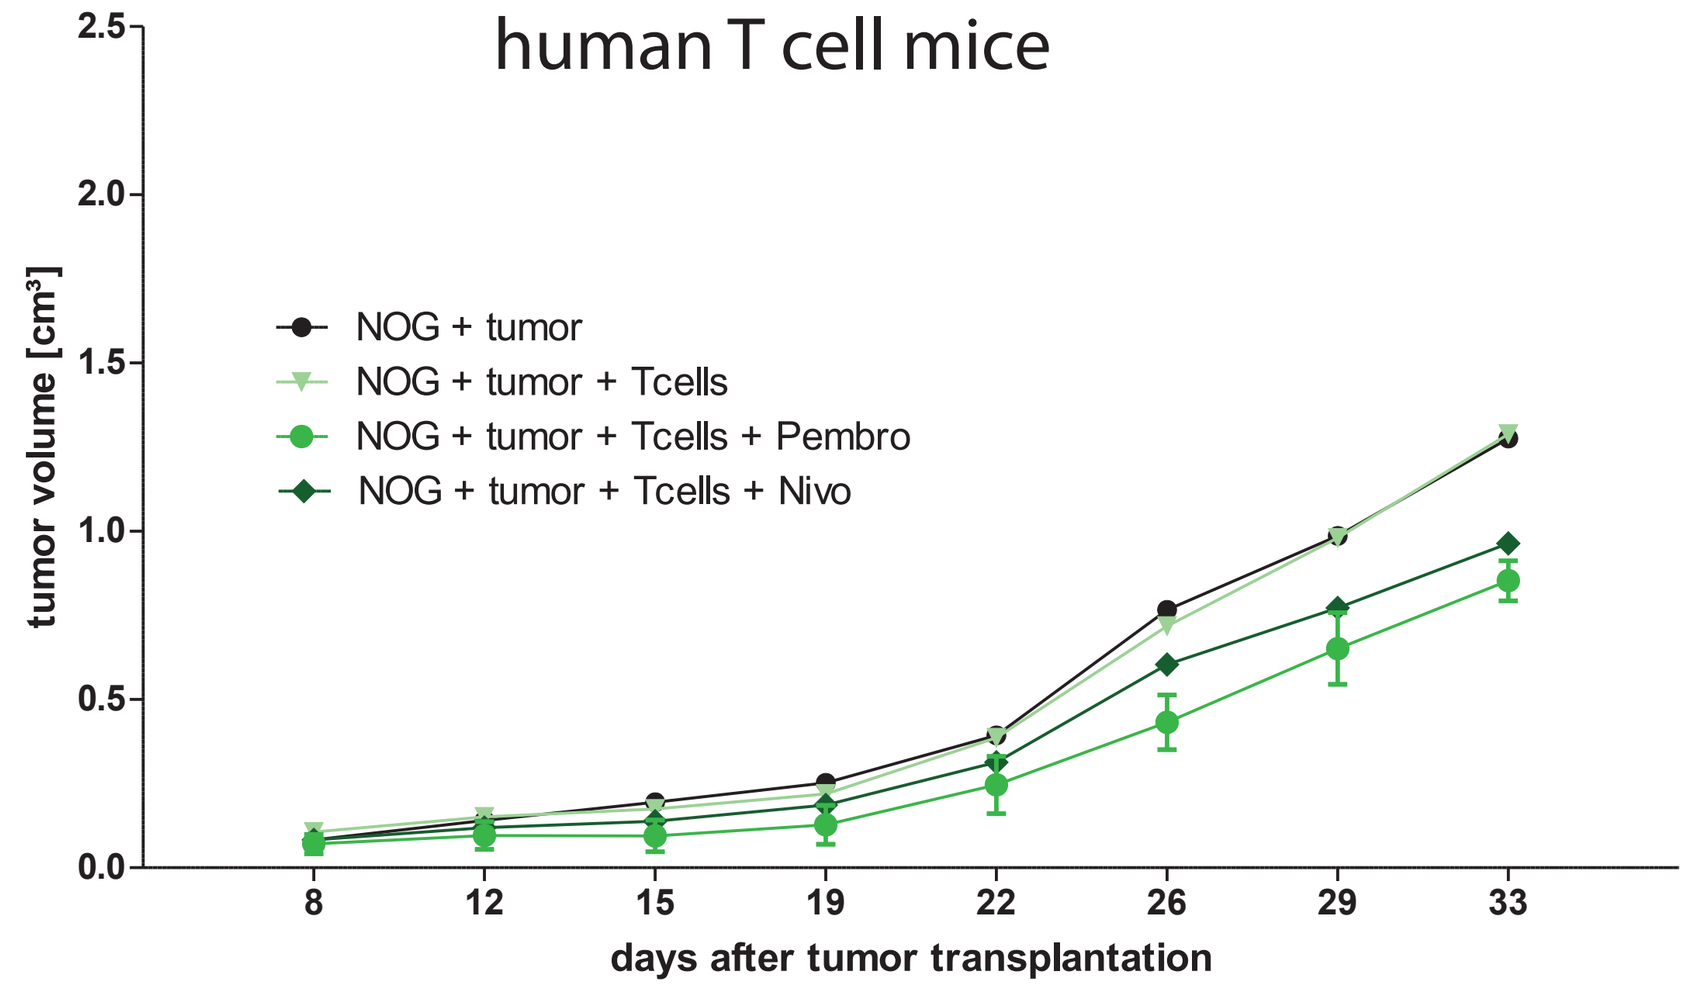
human T cell mice
tumor volume [cm³]
NOG + tumor
NOG + tumor + Tcells
NOG + tumor + Tcells + Pembro
NOG + tumor + Tcells + Nivo
days after tumor transplantation

- comparable growth of Panc12975 on mice with and without T cell humanization, no effects of checkpoint inhibitors

Corresponding address: maria.stecklum@epo-berlin.com; jens.hoffmann@epo-berlin.com

www.epo-berlin.com